Preclinical development of GJB2-GT as a treatment for the autosomal recessive non-syndromic deafness 1A (DFNB1A) using an adeno associated vector-based gene therapy

Christophe Tran Van Ba[1], Guillaume Olivier[1], Sandra Pierredon[1], Anne-Valérie Héritier[2], Charlène Vaux[1], Amrit Singh-Estivalet[2], Charlène Joséphine[1], Andrea Lelli[2], Solène Roux[2], Lise Barrot[1], Julie Duron Dos Reis[1], Pierre Rambeau[1], Anais Pages[1], Laurène Heriaud, Pauline Liaudet[1], Muriel Sudres[2], Nicolas Michalski[2], Rafik Boudra[1], Arnaud Giese[1], Christine Petit[2,3], Laurent Desire[1]

[1]Sensorion, [2]Institut de l'Audition, [3]Collège de France

Abstract

In the world, the estimated prevalence of severe or profound deafness in human is 1 out of 1000 neonates, and genetic factors account for half of the cases. Pathogenic variants of GJB2, the gene encoding for Connexin 26, are involved in 50% of congenital deafness and are mostly associated with an autosomal recessive non-syndromic DFNB1A. In the cochlea, GJB2 is largely expressed in the supporting cells (SCs) of the sensory epithelium, fibrocytes, and basal and intermediate cells of stria vascularis but not in sensory hair cells. It is hypothesized that Cx26 is essential for the recycling of potassium, which is essential for the proper functioning of sensory hair cells, but in vivo studies also suggest that Cx26 deficiency leads to cochlear developmental disorders. Gene therapy is a promising therapeutic strategy for autosomal recessive forms of deafness, and Adeno-Associated Vectors (AAVs) are being developed to this aim. Here, we have developed GJB2-GT, an Adeno-Associated Virus (AAV) vector for DNFB1A that offers broad coverage of *Gjb2*-expressing cells of the inner ear in both mouse and non-human primates. GJB2-GT was delivered into congenitally deaf conditional *Gjb2* mutant mouse ears through the round window (RW). Intracochlear injections of GJB2-GT into conditional *Gjb2*-mutant mouse inner ears lead to improvement of hearing thresholds as early as 3 weeks post-injection in a dose dependent manner. Efficacy on on-going cohorts, dose-response experiments, early biodistribution and toxicology studies in mice are under investigation. In parallel, GJB2-GT was administered to Non-Human Primates (NHP) using the surgery and device used in human. Early tolerability and biodistribution of GJB2-GT studies were conducted in both species. Three weeks post-surgery, ABR measurements and DPOAE amplitudes remained contained within the normal hearing threshold range of NHPs indicating that GJB2-GT was well tolerated. Whole mounts and cryosections of injected inner ears were analyzed to assess the AAV tropism. For both products, a vast majority of SCs that naturally express *GJB2*, including the great epithelial ridge cells, the lateral epithelial ridge cells, border cells, phalangeal cells, pillar cells, fibrocytes of the lateral wall and spiral limbus were transduced along the tonotopic axis. No transduction was found in inner hair cells. GJB2-GT allows to efficiently and safely target the cells that naturally express GJB2 in the cochlea with both tropism and levels compatible with therapeutic intervention in human. These data support GJB2-GT development and constitutes a major step toward our future clinical trials to restore physiological hearing in DFNB1A patients.

1. A new conditional mouse model to study DFNB1A physiopathology: Gjb2$^{cKO1/cKO1}$

Natural history of Gjb2$^{cKO1/cKO1}$

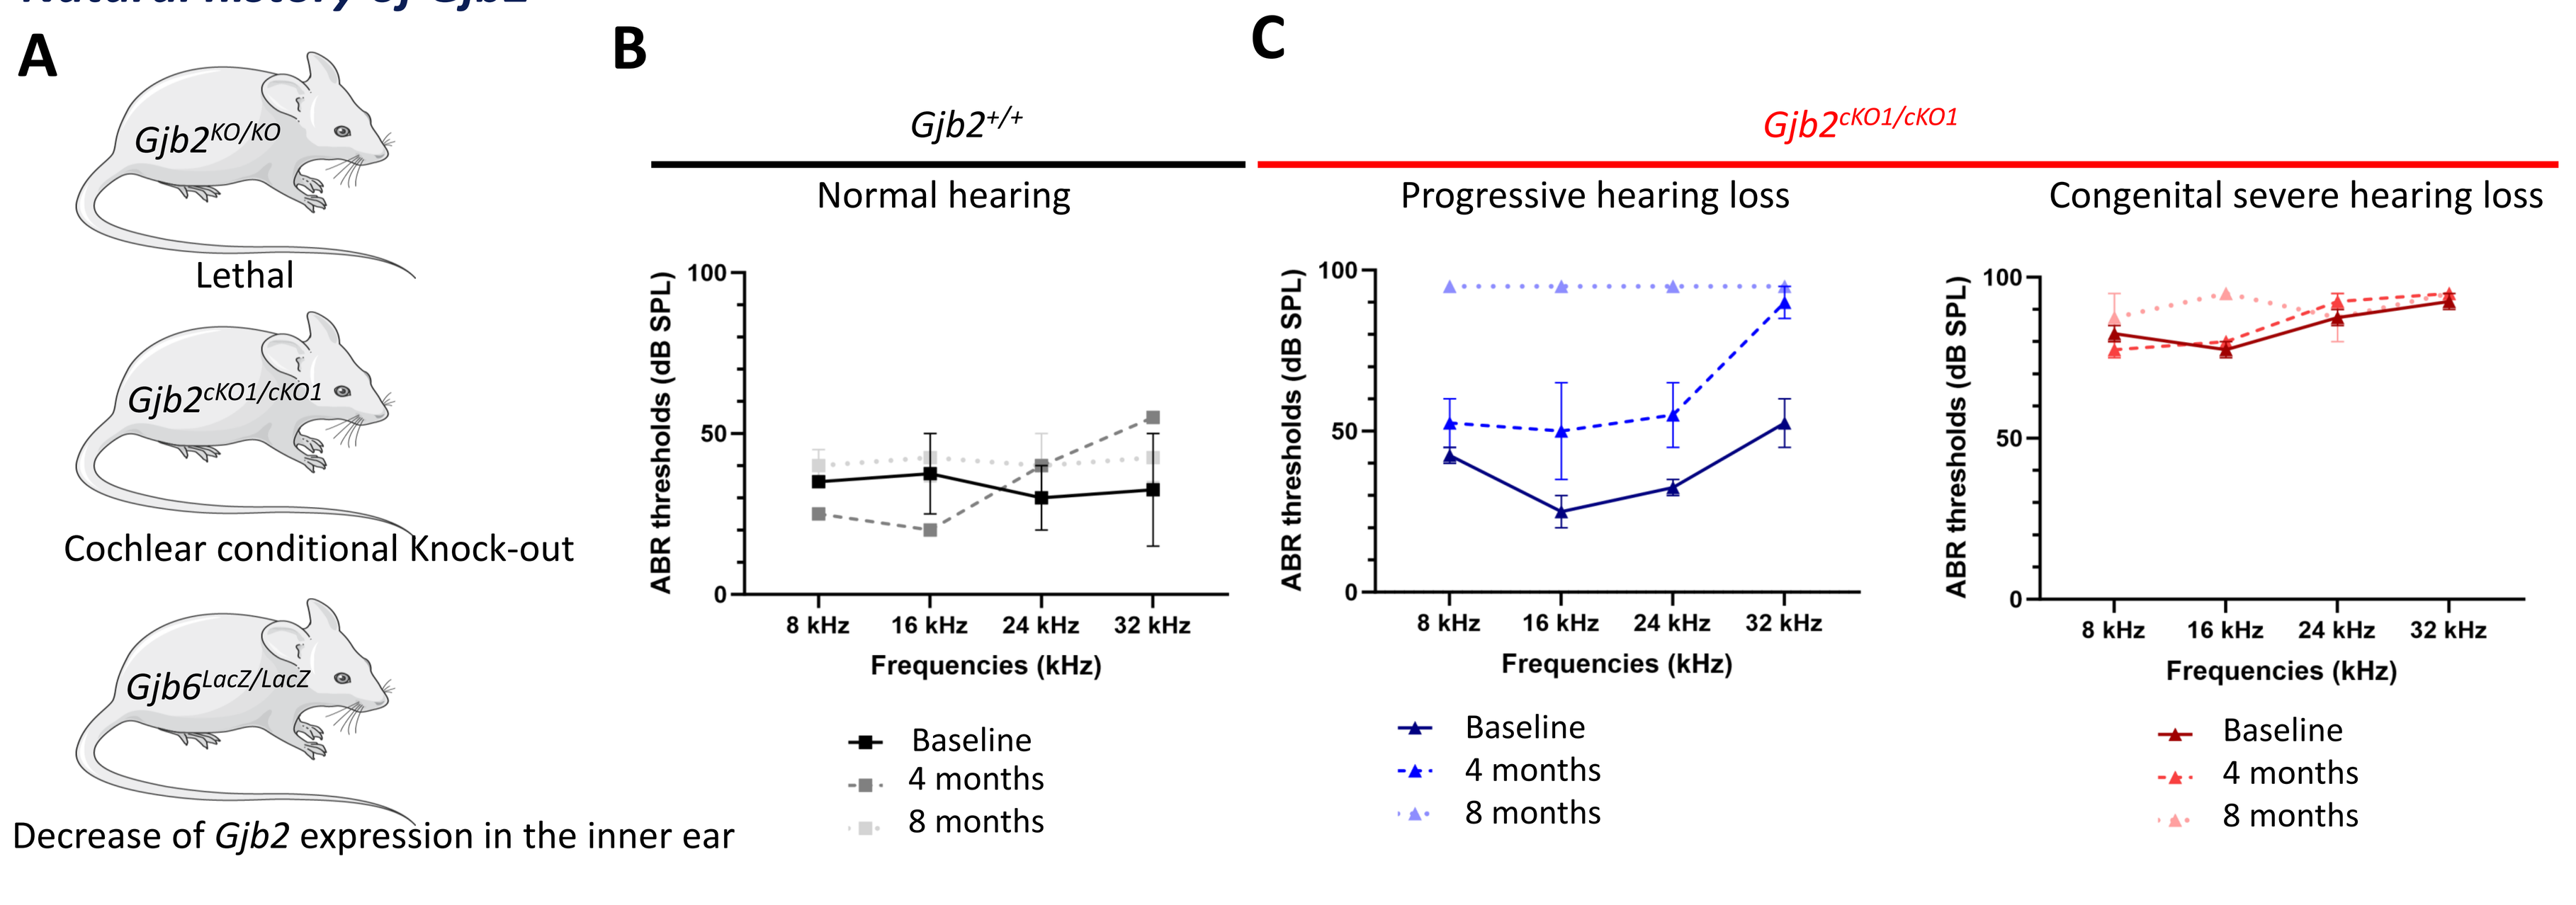
A
Gjb2KO/KO
Lethal
Gjb2cKO1/cKO1
Cochlear conditional Knock-out
Gjb6LacZ/LacZ
Decrease of Gjb2 expression in the inner ear
B
Gjb2+/+
Normal hearing
C
Gjb2cKO1/cKO1
Progressive hearing loss
Congenital severe hearing loss
ABR thresholds (dB SPL)
Frequencies (kHz)
8 kHz
16 kHz
24 kHz
32 kHz
Baseline
4 months
8 months

Figure1: A. *Gjb2$^{cKO1/cKO1}$* mutant mouse model is a useful tool to study DFNB1A physiopathology and to perform proof of concept (POC) studies since the homozygosity for *Gjb2$^{KO/KO}$* allele is lethal during the embryonic development of the mouse. Moreover, previous reported GJB2 POC studies using *Gjb6* mutant mouse model in which the level of *Gjb2* expression is reduced but still present in the cochlea. **B,C.** Assessment of the hearing function of *Gjb2$^{cKO1/cKO1}$* mutant mice up to 8 months. Control mice from this strain show normal hearing and do not present age related hearing loss due to the absence of Cdh23AHL allele in the genetic background. *Gjb2$^{cKO1/cKO1}$* mutant strain presents two different hearing loss phenotypes: 1/ Progressive mutant mice show hearing loss from 4 months to reach severe/loss hearing loss and 2/Congenital severe hearing loss animals present ABR above 80dB thresholds.

2. Design of Proof Of Concept (POC) studies using Gjb2$^{cKO1/cKO1}$ mutant mice

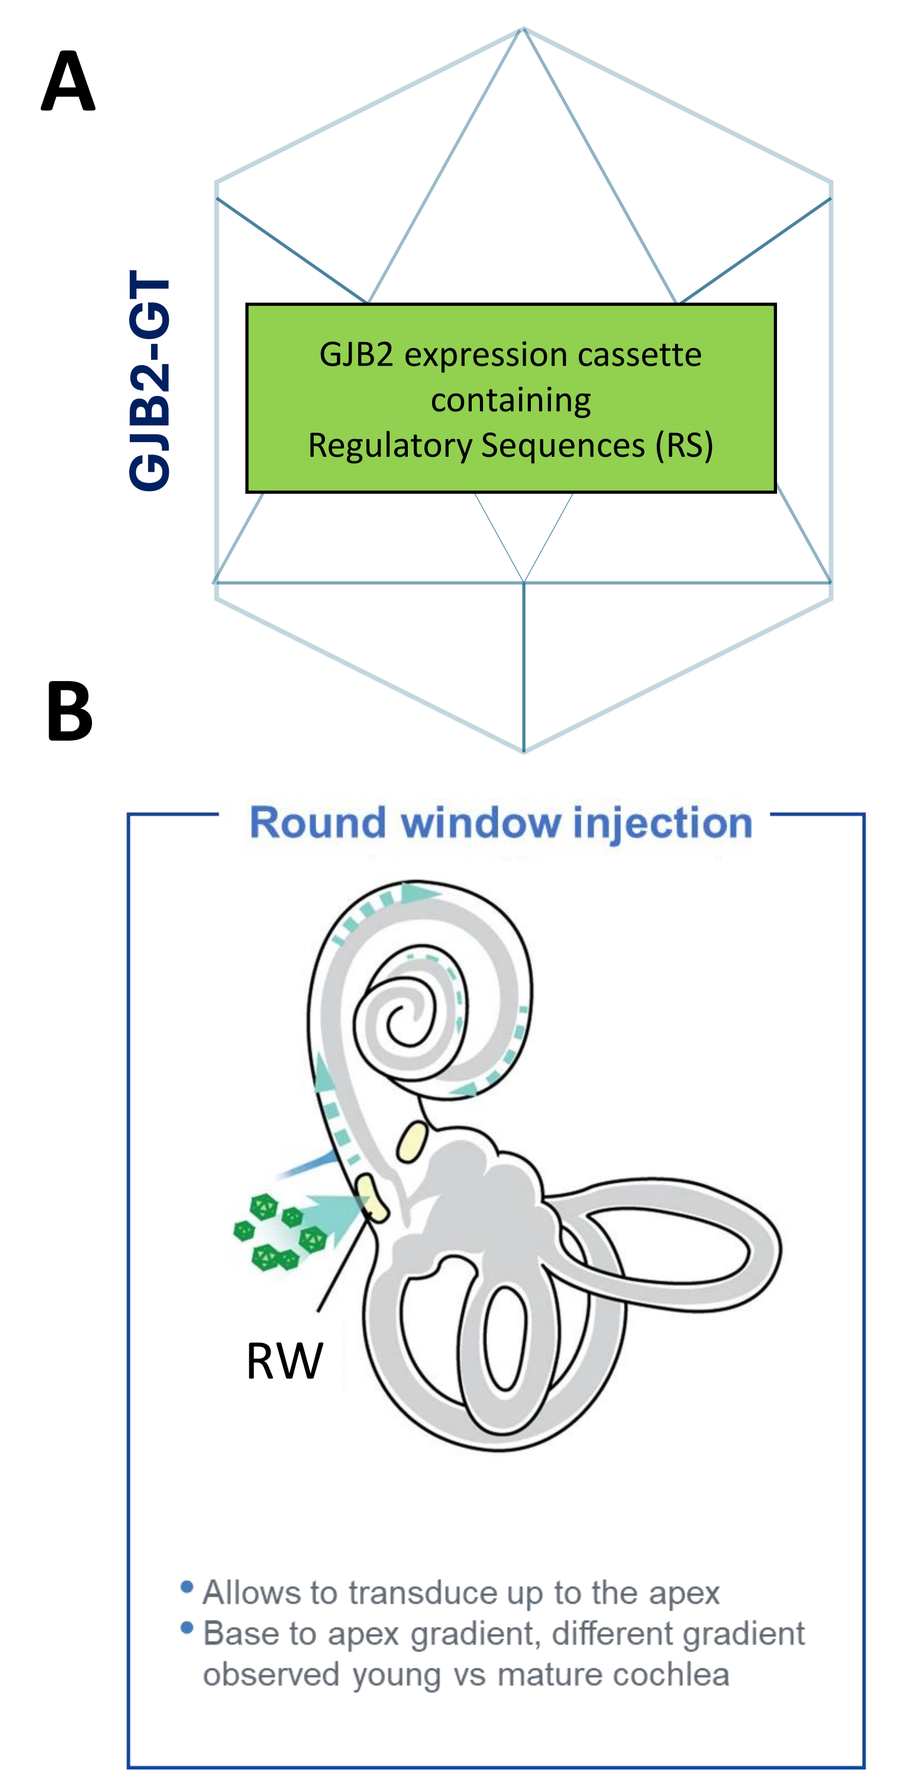
A
GJB2-GT
GJB2 expression cassette containing Regulatory Sequences (RS)
B
Round window injection
RW
Allows to transduce up to the apex
Base to apex gradient, different gradient observed young vs mature cochlea

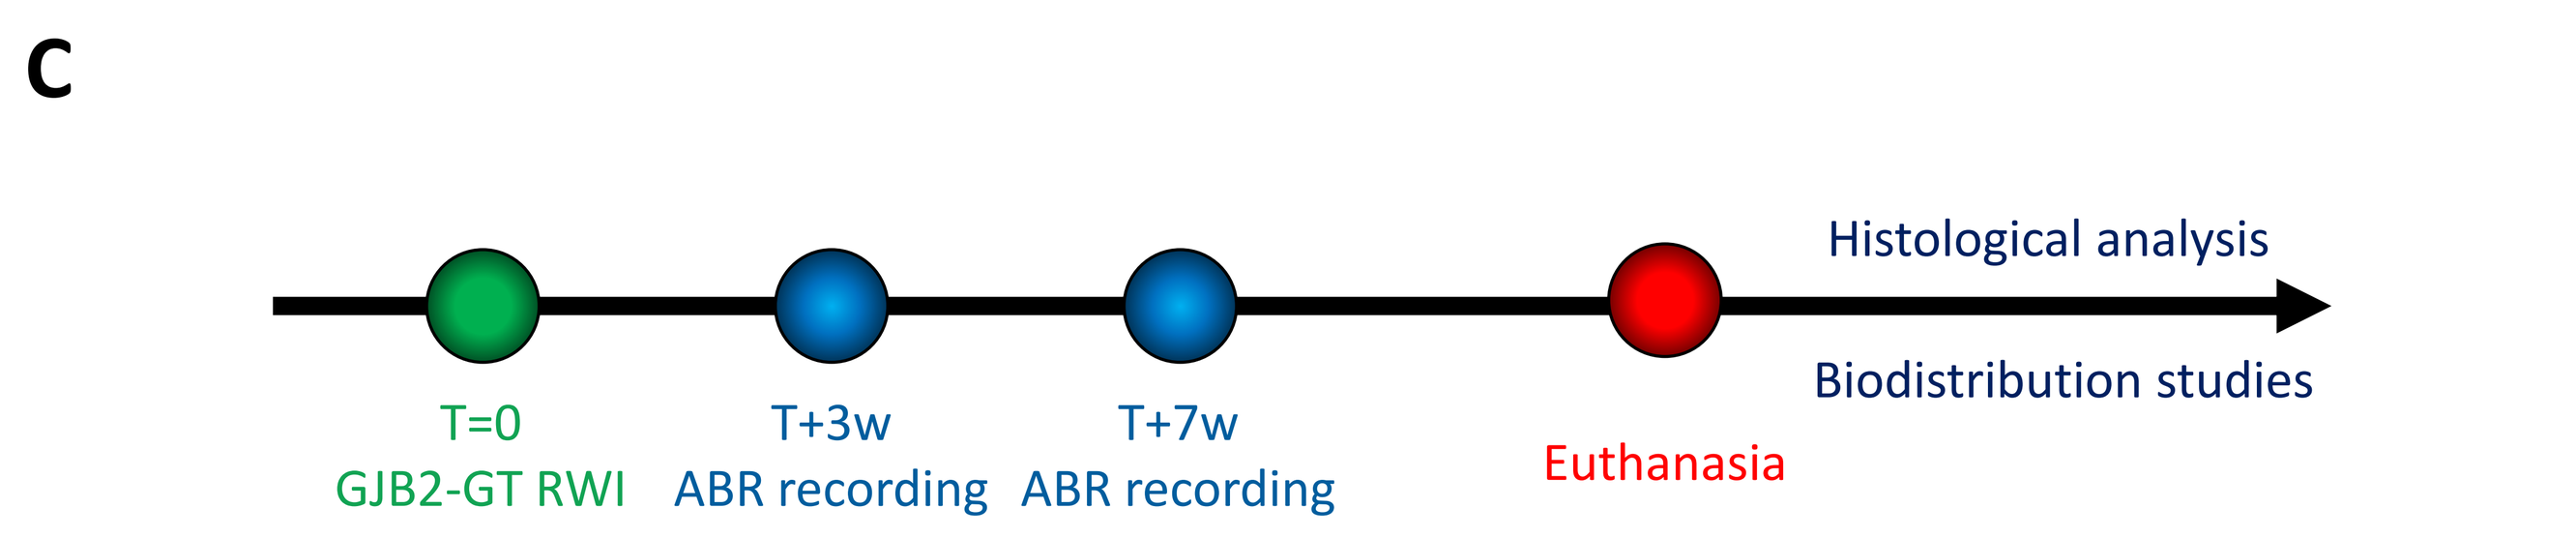
C
T=0
GJB2-GT RWI
T+3w
ABR recording
T+7w
ABR recording
Euthanasia
Histological analysis
Biodistribution studies

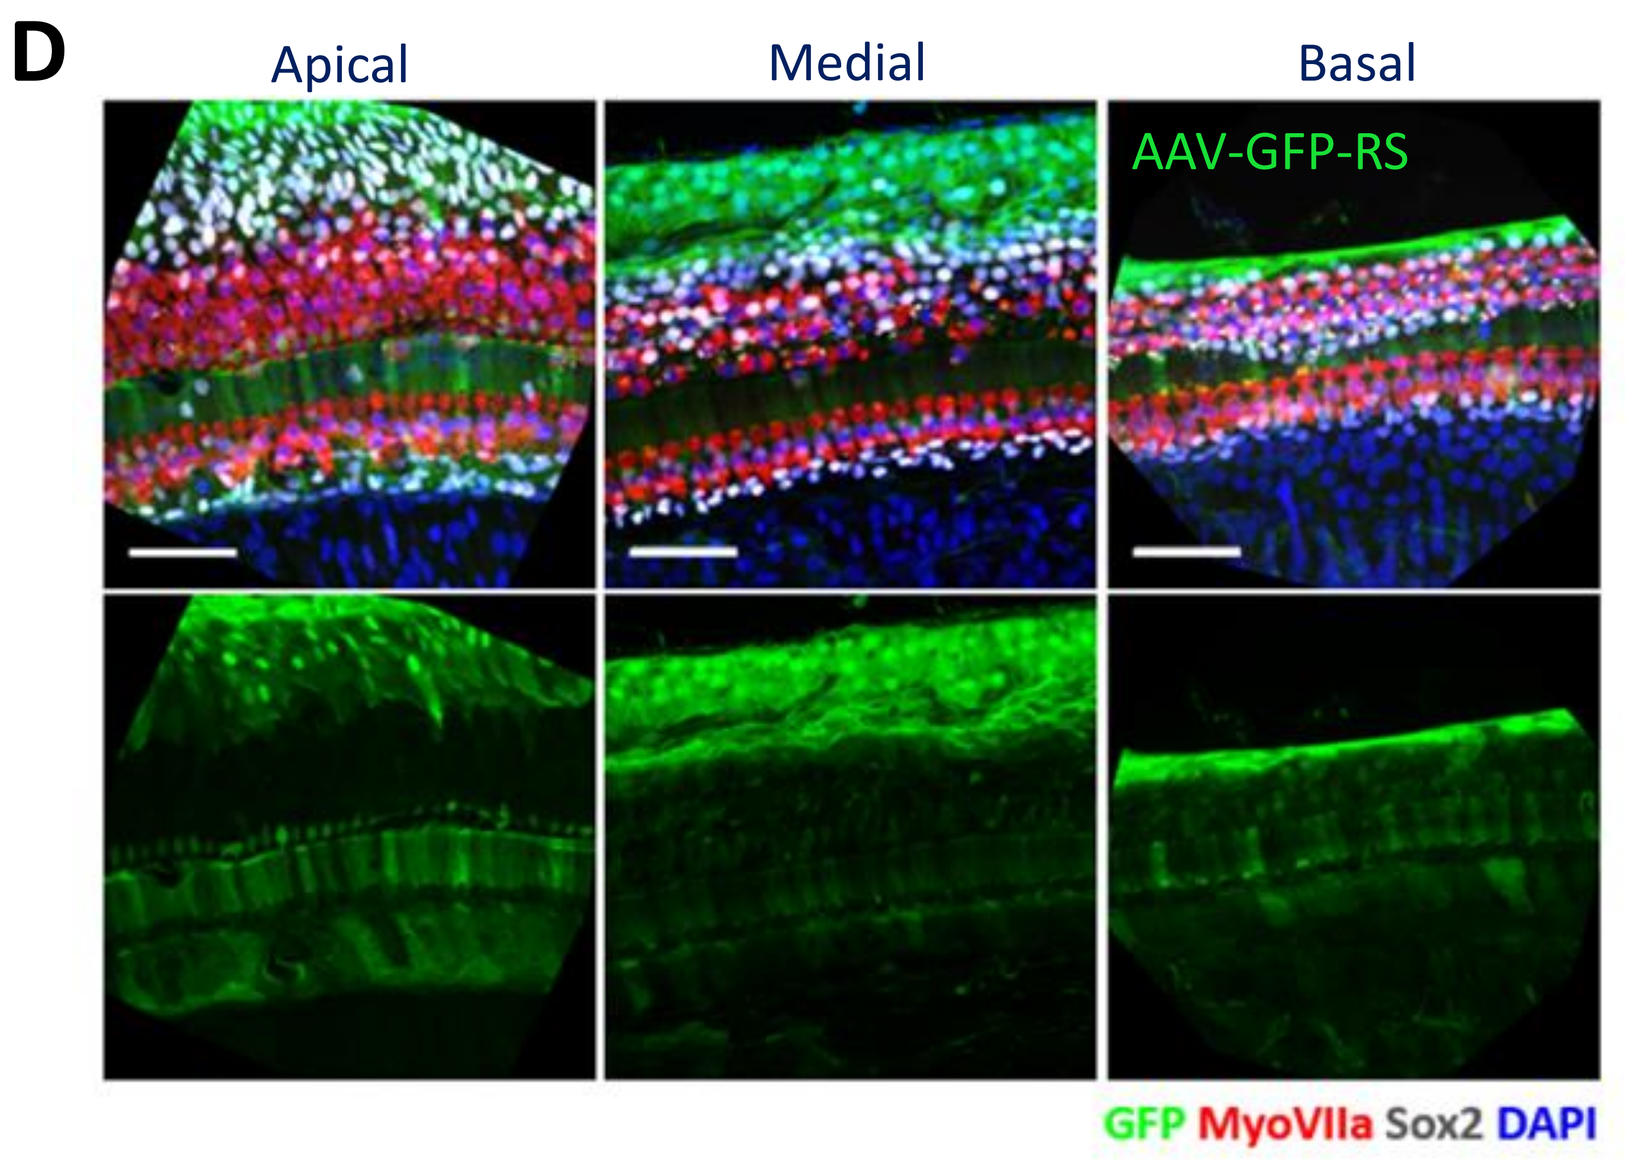
D
Apical
Medial
Basal
AAV-GFP-RS
GFP MyoVIIa Sox2 DAPI

Figure 2: A. Schematic representation of GJB2-GT. The RS is added to prevent transgene expression in sensory hair cells. **B.** Intracochlear injection of GJB2-GT using the round window approach (RWI) in mice. **C.** Design and timeline of proof-of-concept studies. **D.** Whole mount imaging of Organ of Corti from wild-type mice injected with AAV-GFP-RS. Arrows indicate transduced inner hair cells (IHC) Scale bar 40 µm. **The RS efficiently prevents transgene expression in IHC in mice**

3. Efficacy and dose studies using RWI of GJB2-GT in Gjb2$^{cKO1/cKO1}$ mutant mice

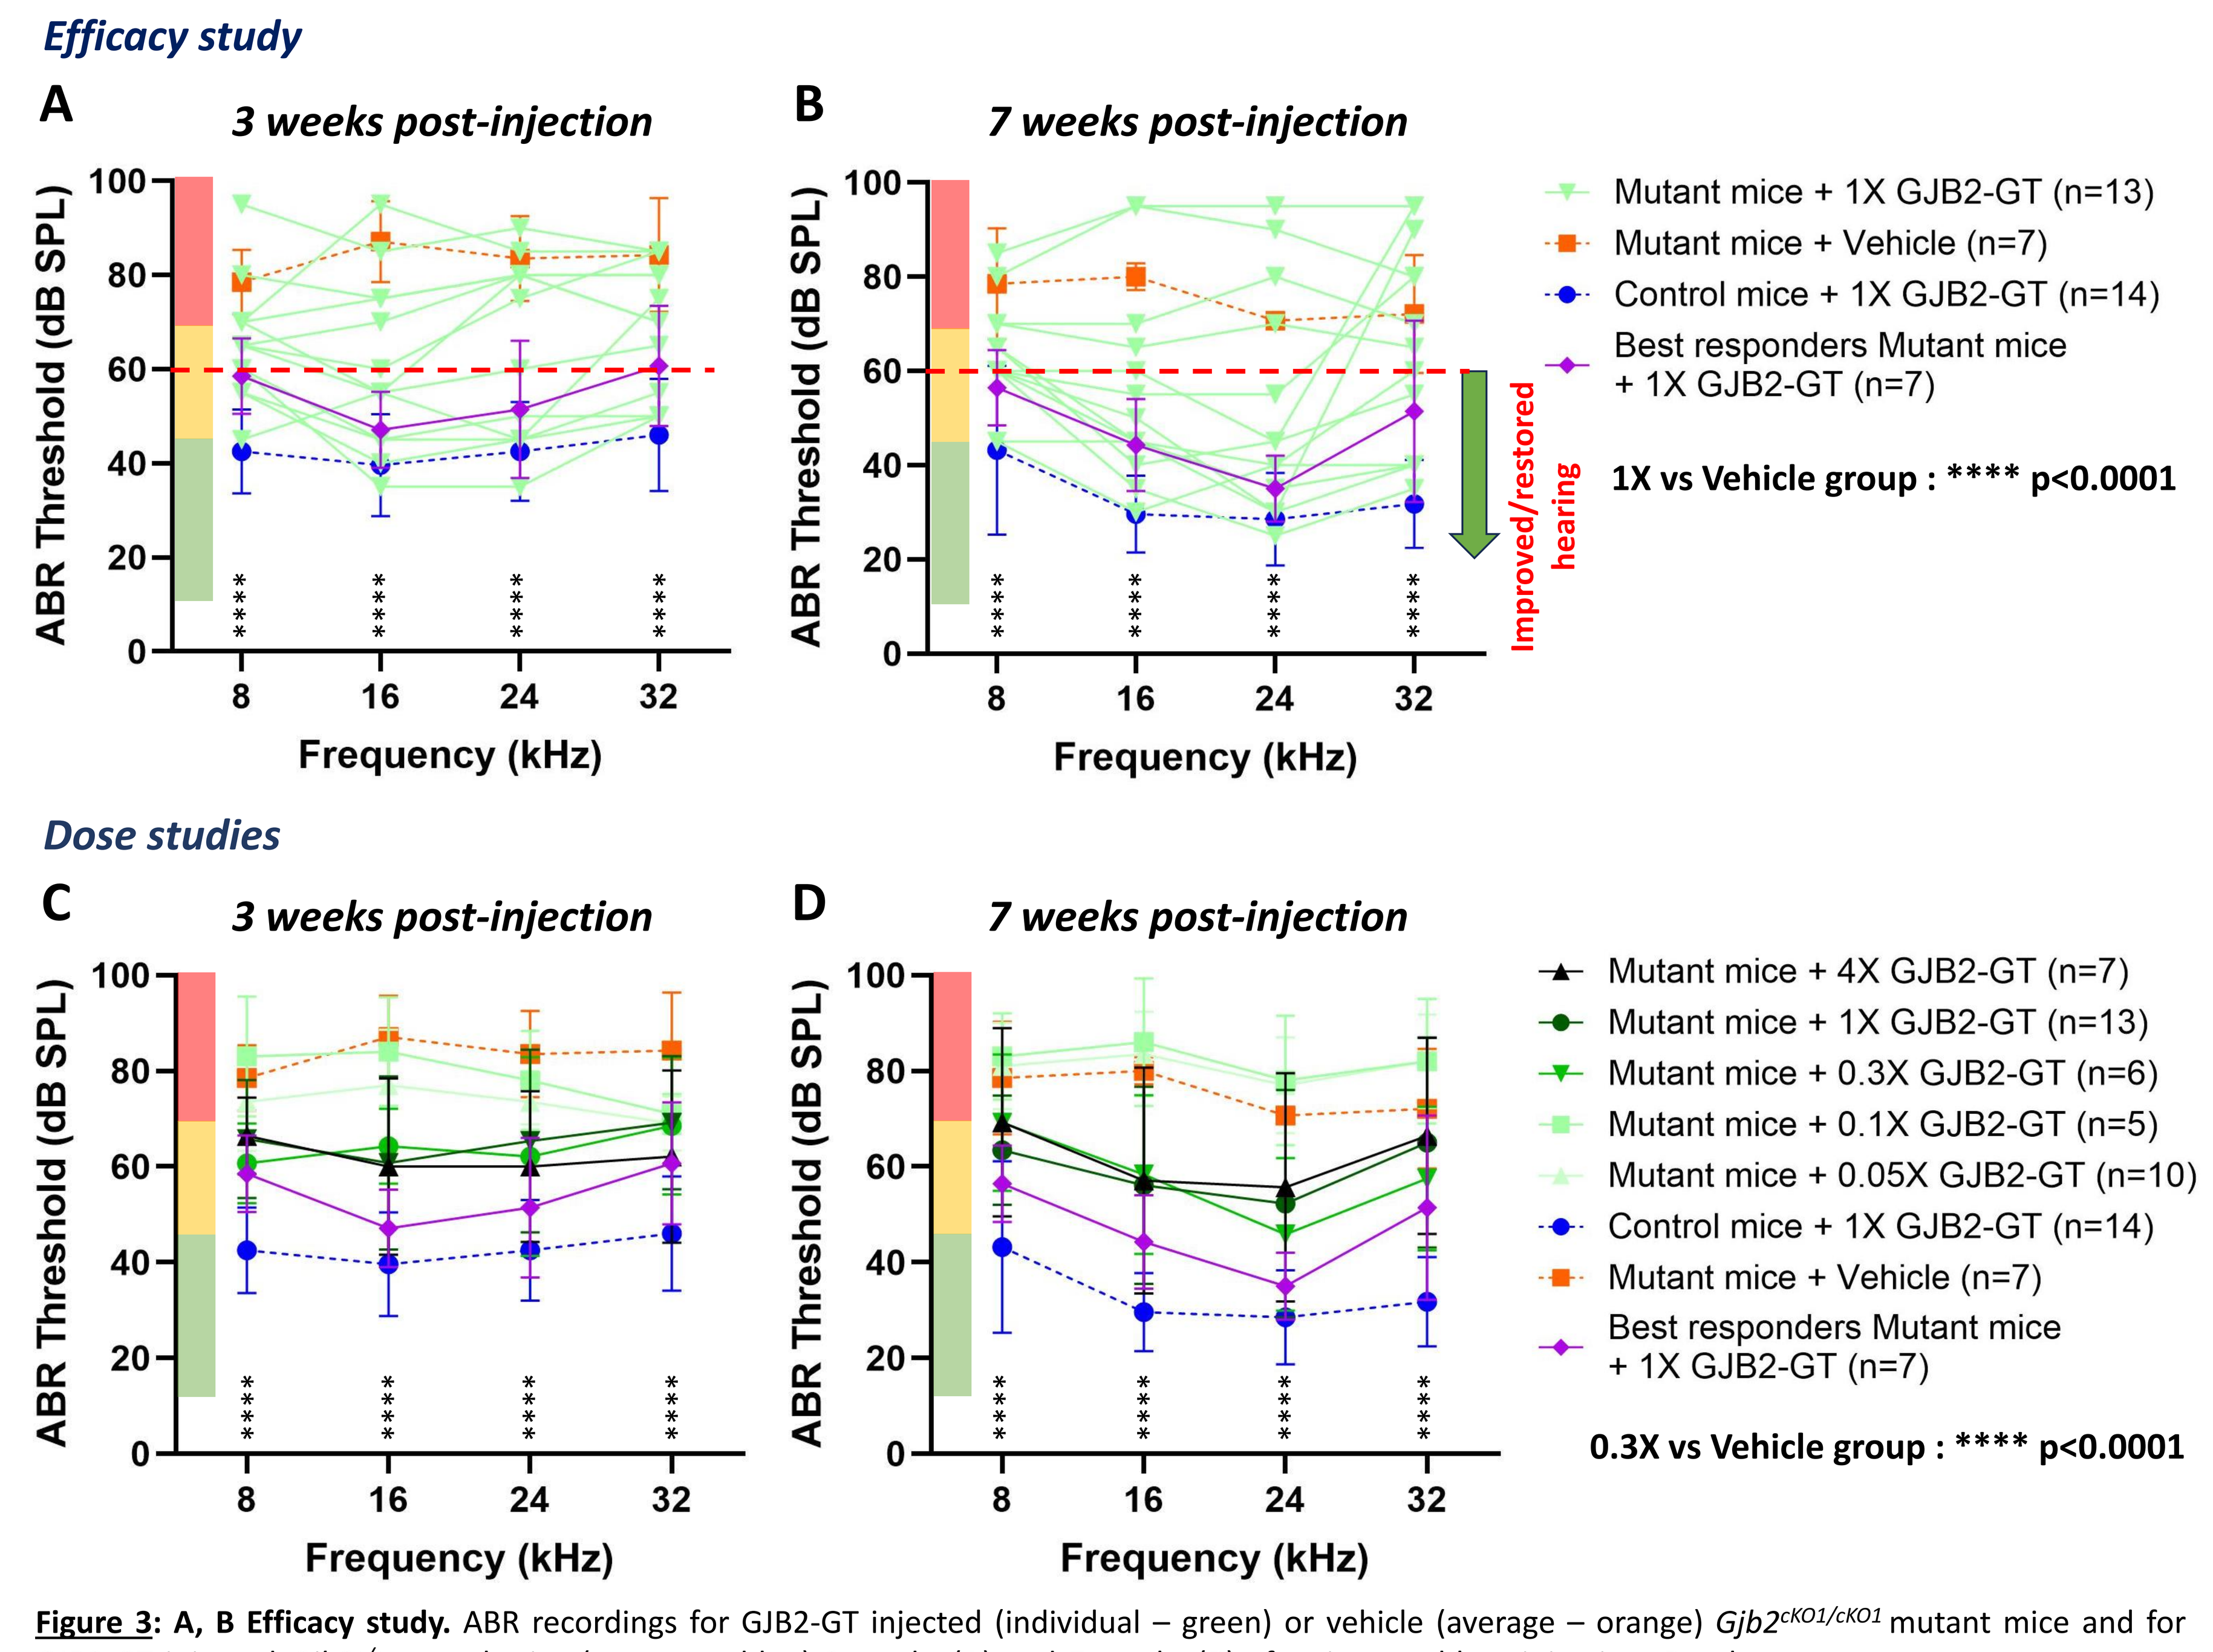
Efficacy study
A
3 weeks post-injection
B
7 weeks post-injection
ABR Threshold (dB SPL)
Frequency (kHz)
Improved/restored hearing
Mutant mice + 1X GJB2-GT (n=13)
Mutant mice + Vehicle (n=7)
Control mice + 1X GJB2-GT (n=14)
Best responders Mutant mice + 1X GJB2-GT (n=7)
1X vs Vehicle group : **** p<0.0001
Dose studies
C
3 weeks post-injection
D
7 weeks post-injection
Mutant mice + 4X GJB2-GT (n=7)
Mutant mice + 1X GJB2-GT (n=13)
Mutant mice + 0.3X GJB2-GT (n=6)
Mutant mice + 0.1X GJB2-GT (n=5)
Mutant mice + 0.05X GJB2-GT (n=10)
Control mice + 1X GJB2-GT (n=14)
Mutant mice + Vehicle (n=7)
Best responders Mutant mice + 1X GJB2-GT (n=7)
0.3X vs Vehicle group : **** p<0.0001

Figure 3: A, B Efficacy study. ABR recordings for GJB2-GT injected (individual – green) or vehicle (average – orange) *Gjb2$^{cKO1/cKO1}$* mutant mice and for GJB2-GT injected *Gjb2$^{+/+}$* control mice (average – blue) 3 weeks (A) and 7 weeks (B) after intracochlear injection. Purple traces represent average ABR measurements for the 7 best responder animals, presenting average ABR thresholds across tested frequencies ≤ 60 dB (red dotted line). Best responder selected animals are the same for both timepoints based on 7 weeks ABR recordings. **GJB2-GT induced a statistically significant hearing recovery for at least 7 weeks after injection in *Gjb2$^{cKO1/cKO1}$* mutant mice.** Colored zones represent approximative profound (>70 dB, red), moderate (45-70 dB, yellow) and no (<45 dB) hearing loss thresholds. **C, D Dose studies.** Average ABR recordings for GJB2-GT 4X to 0.05X injected (black to light green) or vehicle (orange) *Gjb2$^{cKO1/cKO1}$* mutant mice and for GJB2-GT injected *Gjb2$^{+/+}$* control mice (blue) 3 weeks (C) and 7 weeks (D) after injection. Purple traces represent average ABR measurements for the 7 best responder animals, presenting average ABR thresholds across tested frequencies ≤ 60 dB at dose 1X. **A 0.3X dose of GJB2-GT is sufficient to induce a statistically significant hearing recovery for at least 7 weeks after injection in *Gjb2$^{cKO1/cKO1}$* mutant mice.** Colored zones represent approximative profound (>70 dB, red), moderate (45-70 dB, yellow) and no (<45 dB) hearing loss thresholds. **** p<0.0001

4. GJB2-GT transduction of the cochlear neuroepithelium after RW injection in Gjb2$^{cKO1/cKO1}$ mutant mice

Intracochlear injection of GJB2-GT is well tolerated in mice and restores level of Gjb2 expression in the neuroepithelium of the cochlea

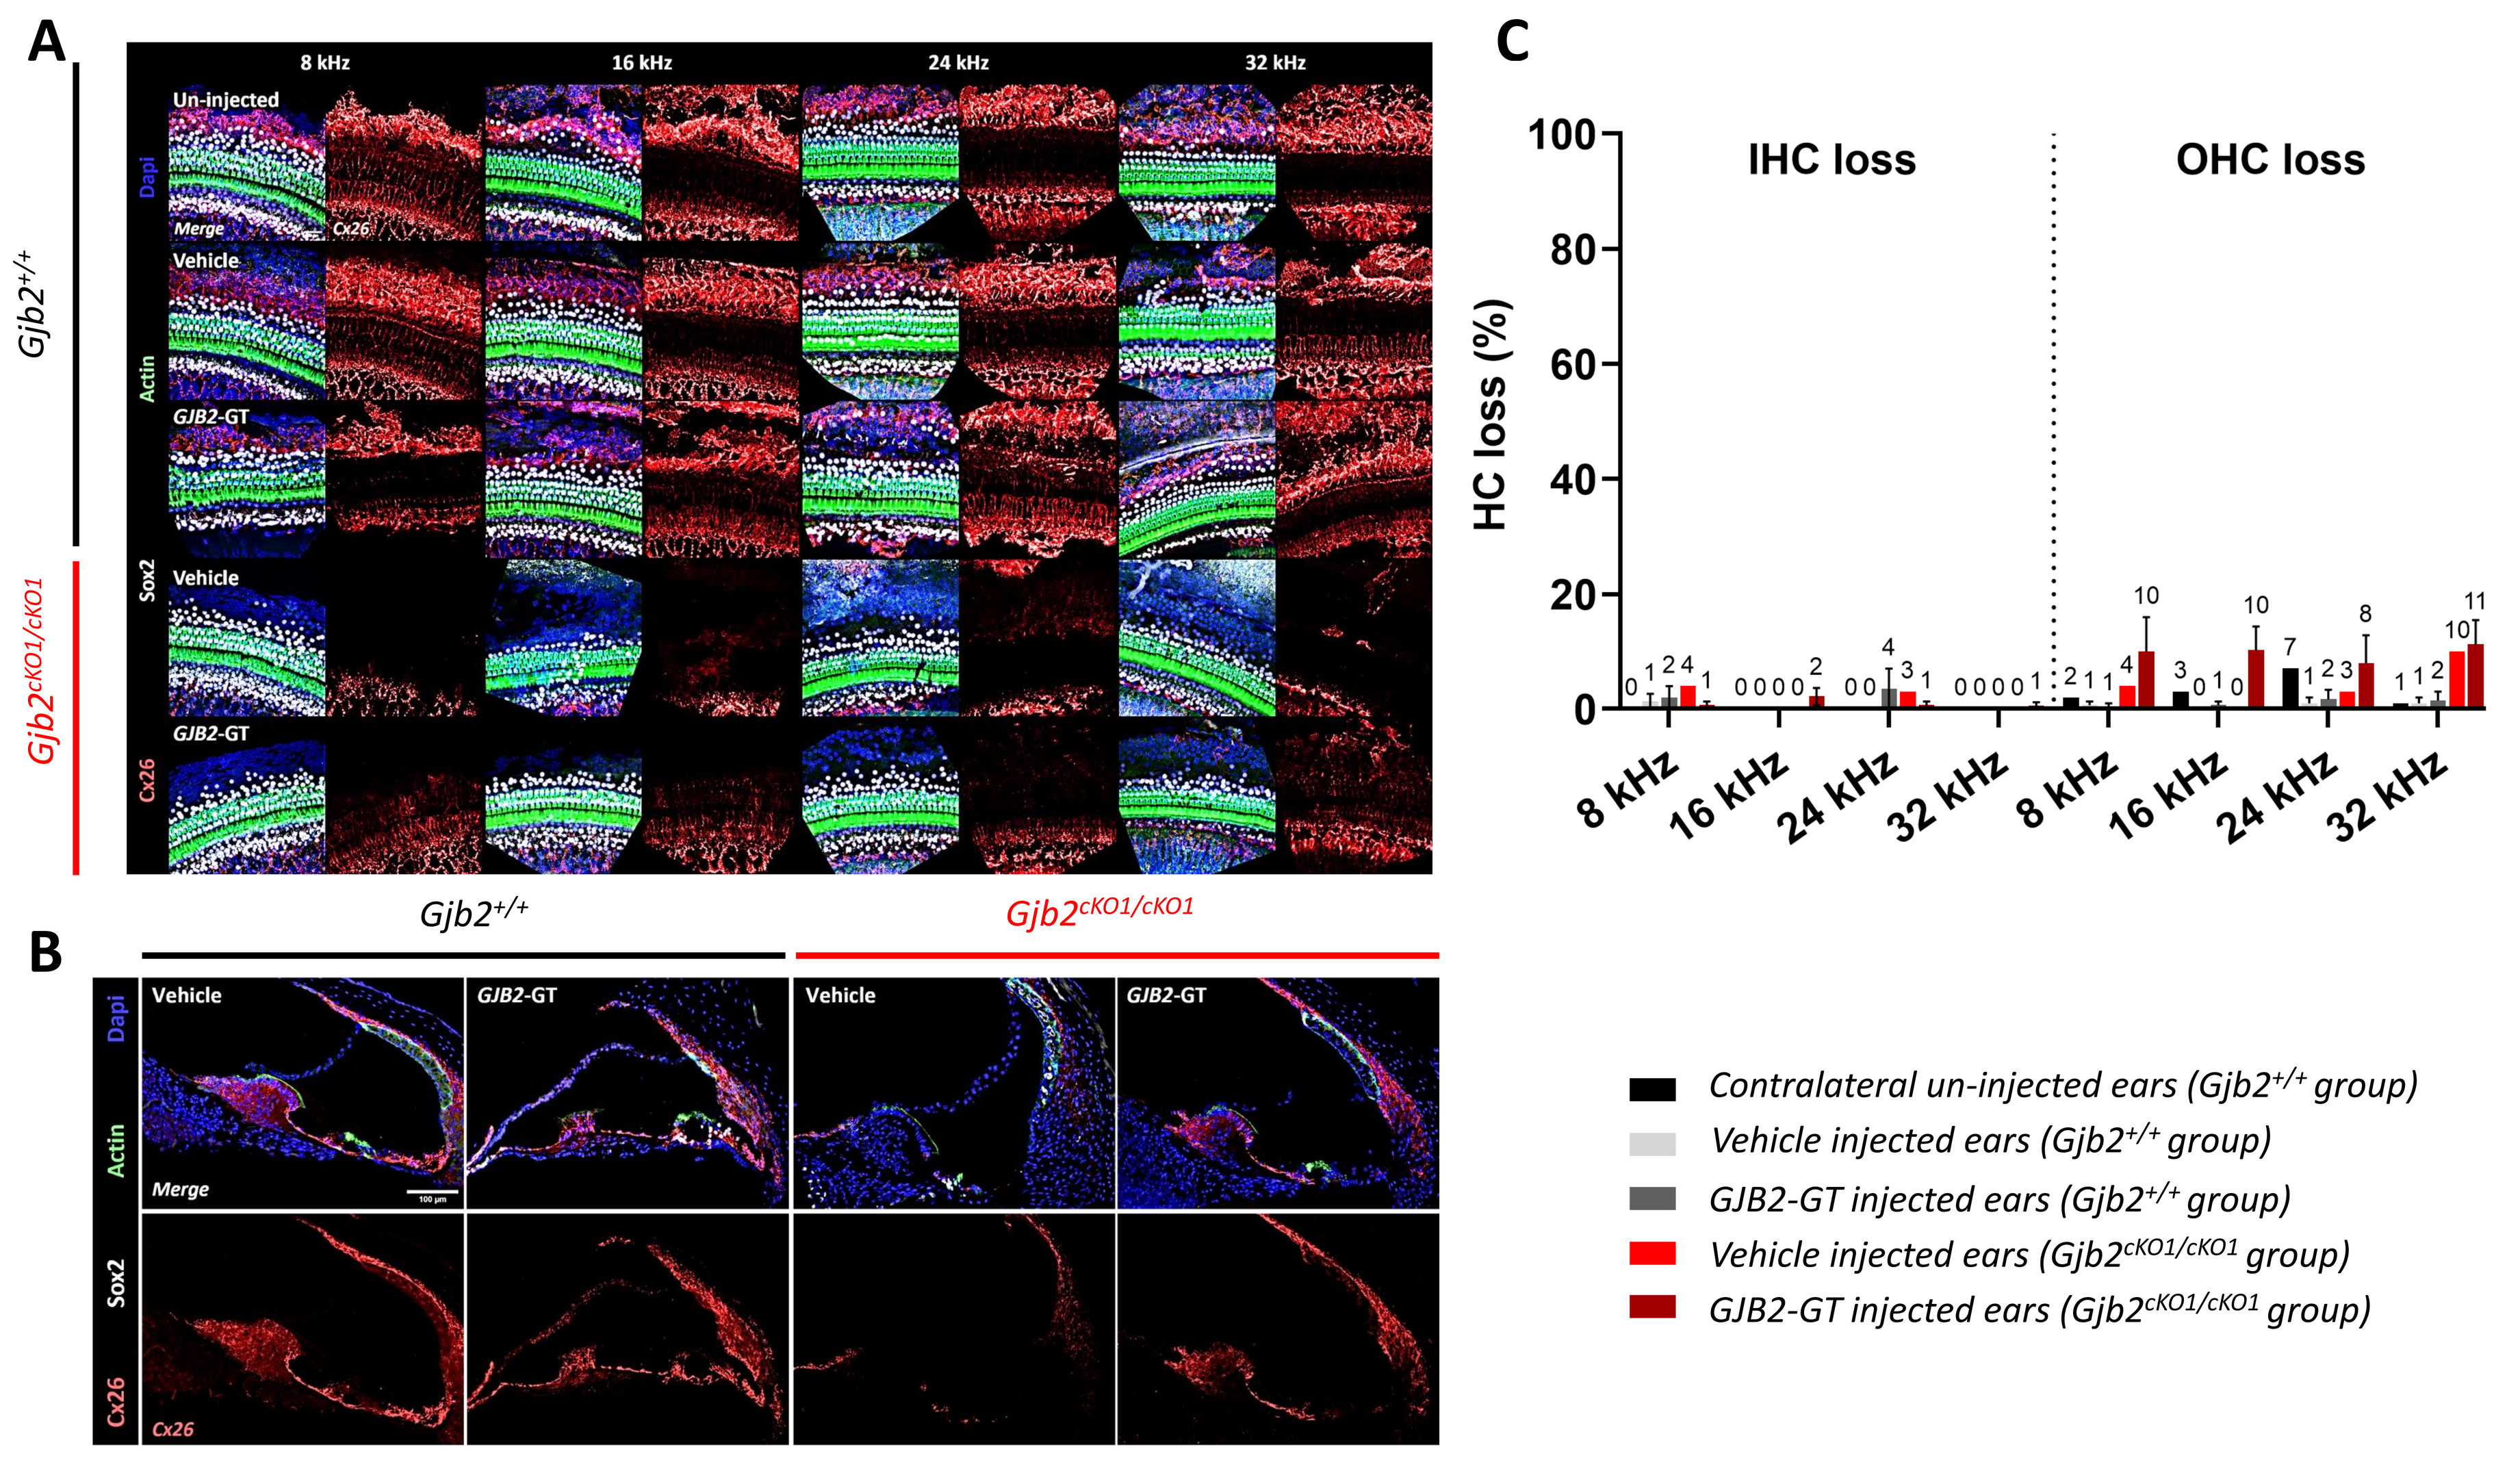
A
8 kHz
16 kHz
24 kHz
32 kHz
Un-injected
Vehicle
GJB2-GT
Dapi
Actin
Sox2
Cx26
Merge
Gjb2+/+
Gjb2cKO1/cKO1
C
IHC loss
OHC loss
HC loss (%)
B
Gjb2+/+
Gjb2cKO1/cKO1
Vehicle
GJB2-GT
Contralateral un-injected ears (Gjb2+/+ group)
Vehicle injected ears (Gjb2+/+ group)
GJB2-GT injected ears (Gjb2+/+ group)
Vehicle injected ears (Gjb2cKO1/cKO1 group)
GJB2-GT injected ears (Gjb2cKO1/cKO1 group)

Figure 4: A, B. Control and *Gjb2$^{cKO1/cKO1}$* mutant mice were injected at P0-P2 in the RW either with vehicle or with GJB2-GT vector at a 1X dose/ear. Three months after injection, histological analyzes were performed on temporal bones (whole mounts at different frequences corresponding to 8, 16, 24 and 32KHz and cryosections). Un-injected animals correspond to the contralateral un-injected ear of animals injected with vehicle. Connexin 26 expression was not impacted by GJB2-GT injection. Intracochlear injection of GJB2-GT restores level of Gjb2 expression in the neuroepithelium of the cochlea of *Gjb2$^{cKO1/cKO1}$* mutant mice. **C.** Quantitative analysis of OHC and IHC loss was performed for each frequency using ImageJ. Numbers indicate the respective % per group and analyzed frequency position. By comparing the values obtained across the various groups, we can conclude that there is no significant loss in the injected groups in relation to the non-injected group at any location in the cochlea.

5. GJB2-GT administration in NHP as currently performed in Sensorion clinical trials

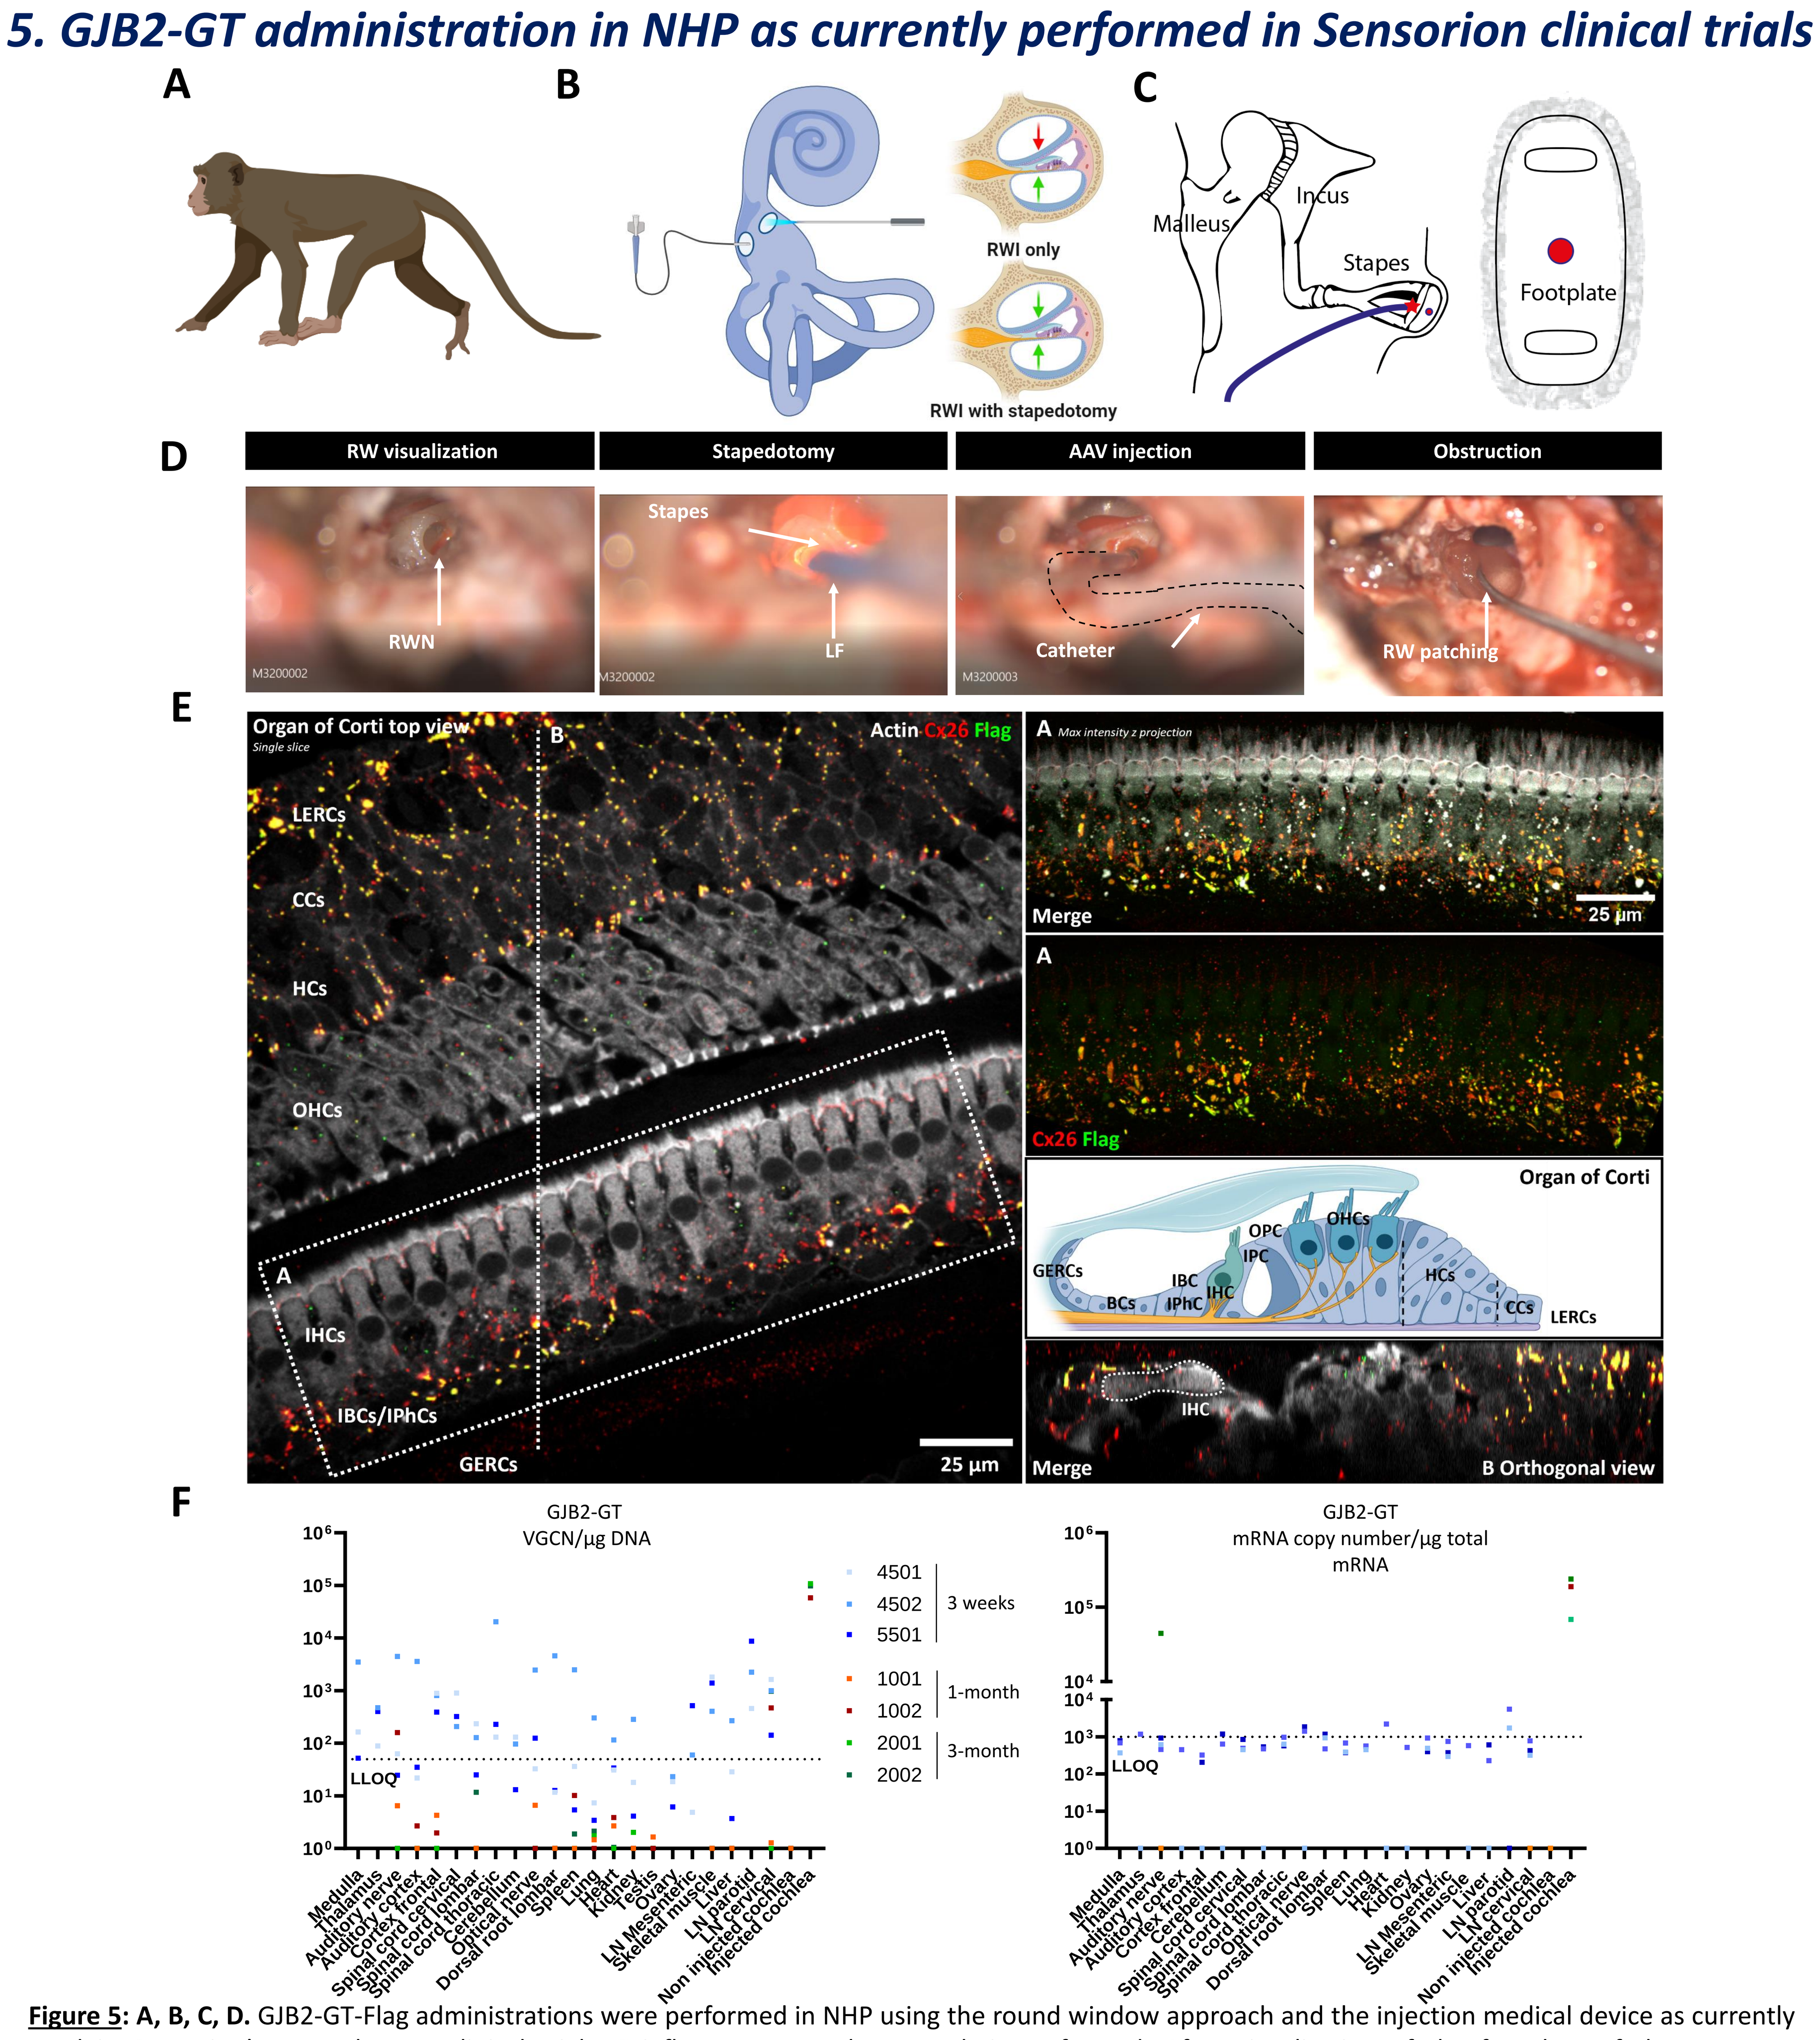
A
B
RWI only
RWI with stapedotomy
C
Malleus
Incus
Stapes
Footplate
D
RW visualization
Stapedotomy
AAV injection
Obstruction
RWN
Stapes
LF
Catheter
RW patching
E
Organ of Corti top view
Single slice
Actin Cx26 Flag
LERCs
CCs
HCs
OHCs
IHCs
IBCs/IPhCs
GERCs
25 µm
Max intensity z projection
Merge
Cx26 Flag
Organ of Corti
GERCs
BCs
IBC
IPhC
IHC
IPC
OPC
OHCs
HCs
CCs
LERCs
B Orthogonal view
F
GJB2-GT
VGCN/µg DNA
LLOQ
4501
4502
5501
3 weeks
1001
1002
1-month
2001
2002
3-month
mRNA copy number/µg total mRNA

Figure 5: A, B, C, D. GJB2-GT-Flag administrations were performed in NHP using the round window approach and the injection medical device as currently used in Sensorion's gene therapy clinical trials. Briefly, a transcanal approach is performed. After visualization of the footplate of the stapes, a stapedotomy is performed and an in-house designed catheter connected to a dedicated injection medical device is inserted in the round window membrane and the intracochlear administration performed. **E.** Three weeks post-injection, NHP were euthanized, the temporal bone collected and fixed in PFA. After decalcification, the cochleae were fine-dissected and immunostained. **F.** Biodistribution of vector genome (left panel) and transgene mRNA (right panel) in the cochlea and peripheral tissues of NHP administered with GJB2-GT. The sensitivity of the method allow for accurate quantification of at least 50 copies of vector genomes (VGCN, vector genome copy number) per microgram of genomic DNA, and 10 copies of mRNA in 10 ng of total RNA.

Conclusions

- RW injections of GJB2-GT into conditional *Gjb2*-mutant mouse inner ears lead to improvement of hearing thresholds for at least 7 weeks post-injection in a dose dependent manner.
- GJB2-GT offers broad coverage of *Gjb2*-expressing cells of the inner ear in both mouse and NHPs.
- No transduction was found in inner hair cells, and no hair cell loss or ototoxicity were associated with GJB2-GT.
- GJB2-GT allows to efficiently and safely target the cells that naturally express GJB2 in the cochlea with both tropism and levels compatible with therapeutic intervention in human.
- Biodistribution studies in NHP shows widespread but low level of VCN in peripheral tissues that decreases overtime. Strong detection of vector of DNA and transgene derived mRNA is observed at the injection site (temporal bone) and transgene mRNA is almost undetectable in VCN positive peripheral tissues and contralateral ears, demonstrating very low ectopic Cx26 expression potiential outside the inner ear.
- GJB2-GT constitutes a major step toward our future clinical trials to restore physiological hearing in DFNB1A patients and is progressing into IND/CTA enabling studies with Clinical Trial Applications planned in 2025.